CASE SERIES

Painless Enhanced Autolytic Debridement on 252 Chronic Wounds by Using Polymeric Membrane Dressings*

Dr. Charalambos Agathangelou, PhD in Medical Gerontology St. Demetrianos, Centre of Assisted Living, Rehabilitation and Wound Care, Dhali, Nicosia, CYPRUS

INTRODUCTION

Several types of debridement are available for health care professionals but the decision to debride a patient's wounds lies on the principle that the healing process needs to be improved and accelerated. Many chronic ulcers are covered with slough and/or necrotic tissue, and are painful and challenging to treat because there is no standard protocol that can be applied to all patients. Very often, these kinds of wounds would need to be debrided more than once. We have noticed that after an initial sharp debridement some wounds became again necrotic/sloughy within 24 hours. Optimal and successful wound management needs a treatment plan according to the wound, patients' parameters and available recourses since a fine line exists between beneficial and harmful debridement.

For the past 10 years we have treated these types of ulcers with polymeric membrane dressings* (PMDs) due to their unique properties.

PMDs contain a surfactant, superabsorbent starch and glycerol/glycerin, which work synergistically to promote rapid healing and reduction of inflammation. These dressings also facilitate autolytic debridement by loosening bonds between slough/fibrotic tissue and healthy granulation tissue.

METHOD AND RESULTS

The past 10 years we have treated 252 patients with painful necrotic and/or sloughy wounds where autolytic debridement with PMDs was used. A few cases were partially surgically debrided prior to use of PMDs. PMDs, with and without silver, were applied.

PMDs enhance the autolytic process by reducing the interfacial bonding forces between healthy and necrotic tissue (hence making the necrotic tissue come off easier), and by dissolving dead tissue. Initially the exudates levels increase and daily changes are required to prevent damage to the surrounding skin. Although a slow process, its main advantage over other methods of debridement is that it's painless and encourages the healing process by instigating angiogenesis. Of the 252 cases, 25% were partially surgically debrided prior to use of PMDs. 9% were surgically debrided after PMDs (moistened with a few drops of sterile saline or water) were applied to the wound in order to soften the necrotic tissues.

Debridement took between 3 and10 days, depending on type and size of the wound. The approach resulted in a smaller wound to be closed than would have been created by typical aggressive surgical debridement.

	Total	Only PMD	Surgically prior to PMD	Surgically after first moistened by PMD
Arterial wounds	26	26		
Venous ulcers	35	35		
Diabetic ulcers	55	17	38	
Heel pressure ulcers	62	37	25	
Pressure ulcers	74	52		22

DISCUSSION

At our Clinic we focus on continuous debridement, exudate control, prevention of wound trauma and infection as well as pain control. We also take into account changes in the skin such as appearance, structure, mechanical properties and barrier function due to ageing. PMDs help us achieve our goals; as a single treatment modality it's effective for all phases of wound healing and doesn't require additional treatments or additives. This made it especially helpful in settings where patients or their family had to perform dressing changes at home.

One reason for primarily choosing autolytic debridement was that surgical intervention was often too stressful for patients with low albumin, hemoglobin and iron levels. Enhanced autolytic debridement is a significant advantage over other methods of autolytic debridement in that it's painless and simultaneously promotes the healing process by instigating angiogenesis. PMDs helped us to provide continuous debridement, exudate control, prevention of wound trauma, prevention of infection and pain control.

As it is impossible to show all cases here I am presenting a representative selection of cases to show how the PMDs work and support our initial decision of using it as a single modality treatment.

An 83 year old patient with Type 1 Diabetes, Peripheral Arterial Disease and Dialysis was referred to our clinic for palliative care. He had a painful wound on his ankle that had been open for 9 months. Angioplasty could not be performed due to his medical condition. He complained over extreme pain and had suffered many sleepless nights. Wet to dry gauze was initially used to debride the wound with no success.

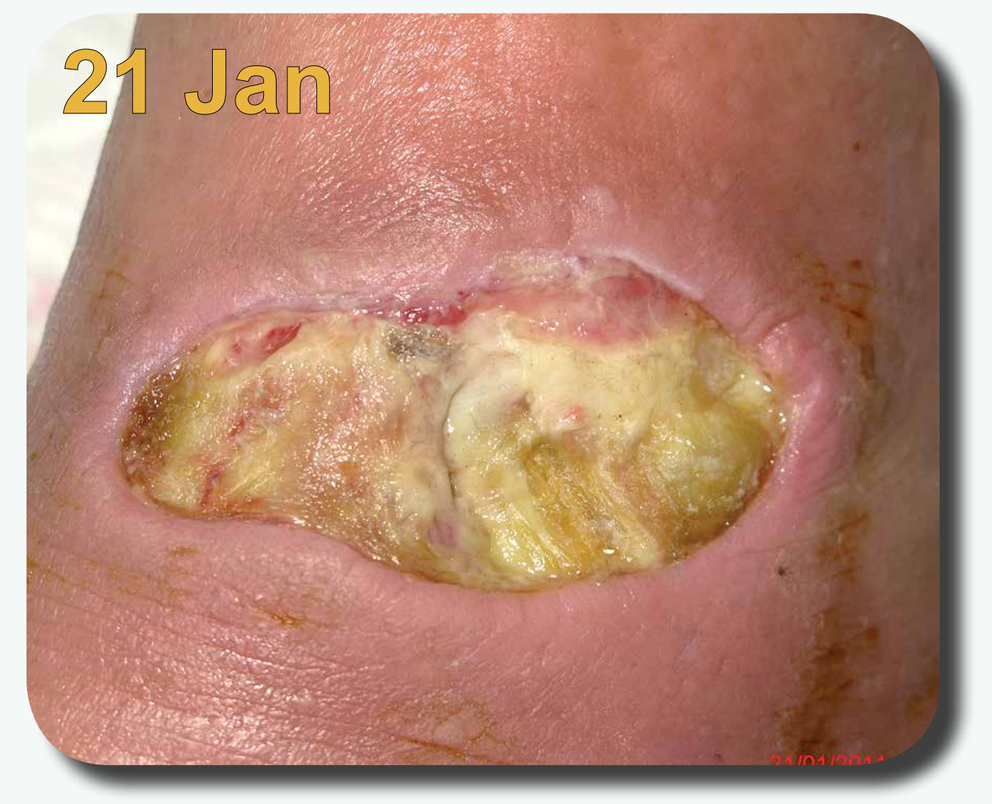
21 Jan

On assessment, the wound is a painful, 10 out of 10, covered with yellow slough which could not be removed by surgical debridement due to an arterial occlusion. PMDs were used in order to alleviate pain and instigate autolytic debridement. Analgesia was given every 4 hours.

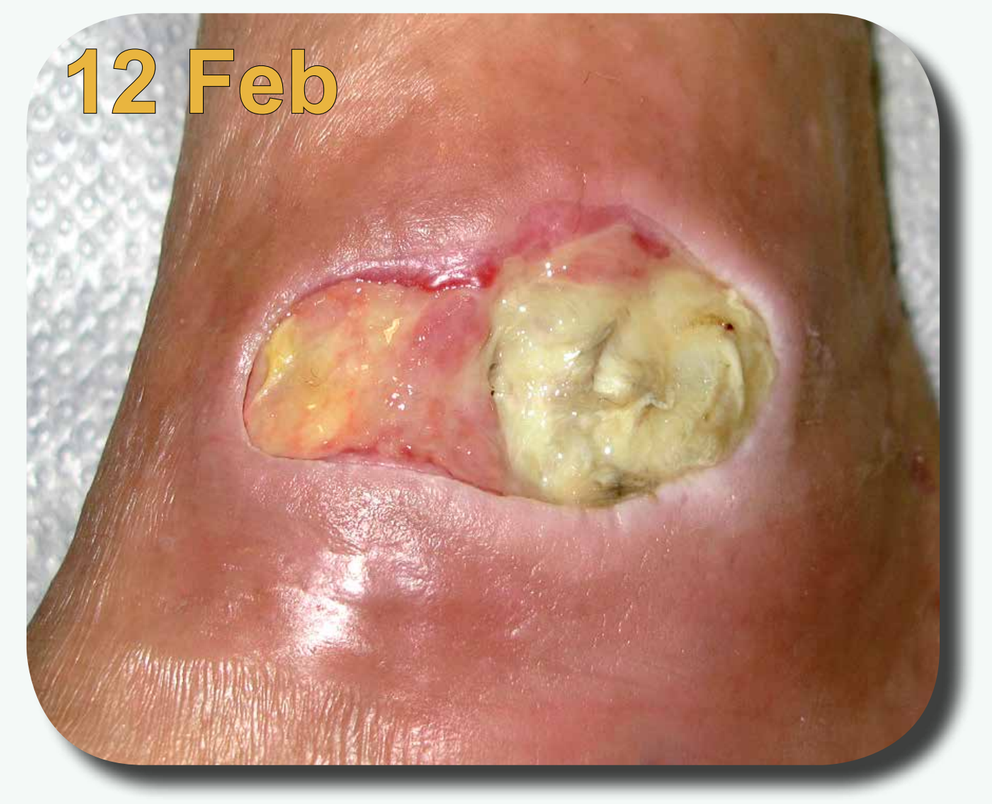
12 Feb

Pain level reduced from 10 to 6 probably due to PMDs capability to absorb sodium ions, from the skin and the subcutaneous tissues resulting in reduced nociceptor conduction, hence alleviating pain. The surfactant dissolves debris by loosening the bonds between the necrotic tissues and absorbing the slough into the dressing. Analgesia was given only at night.

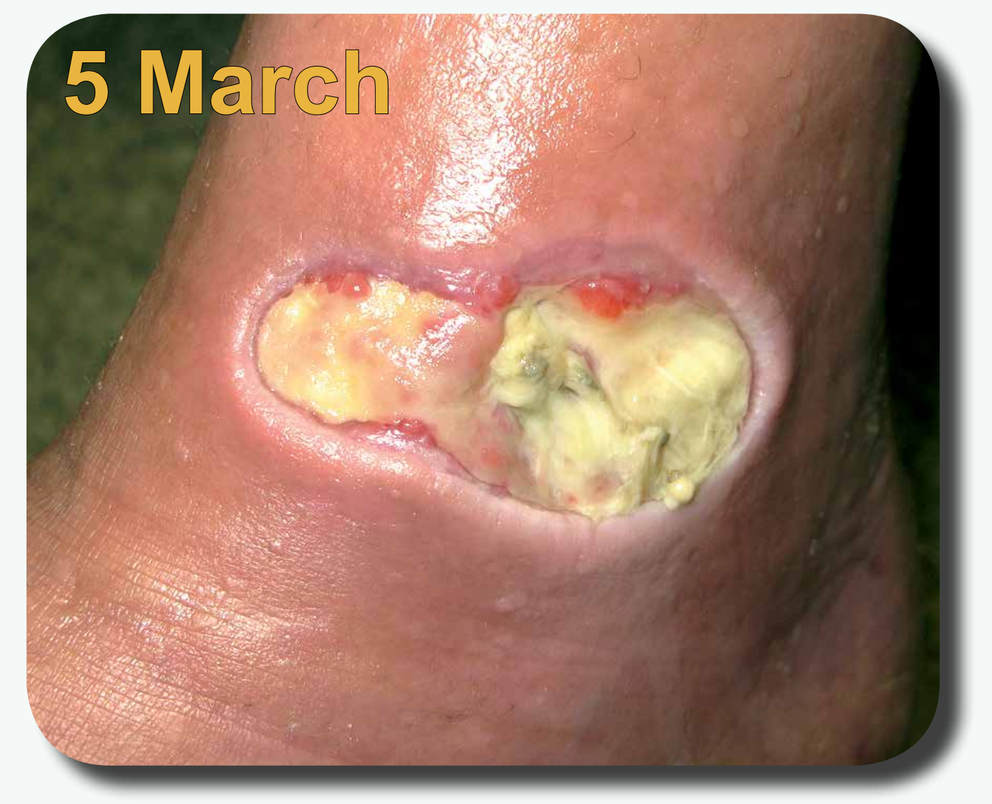
5 March

Reinforced autolytic debridement takes place constantly throughout the healing process, clearing the wound of necrotic tissue during dressing wear time. Pain level dropped to zero and no analgesia was needed any more. The dressing does not adhere to the new tissue and therefore does not cause any secondary damage to the wound at dressing changes.

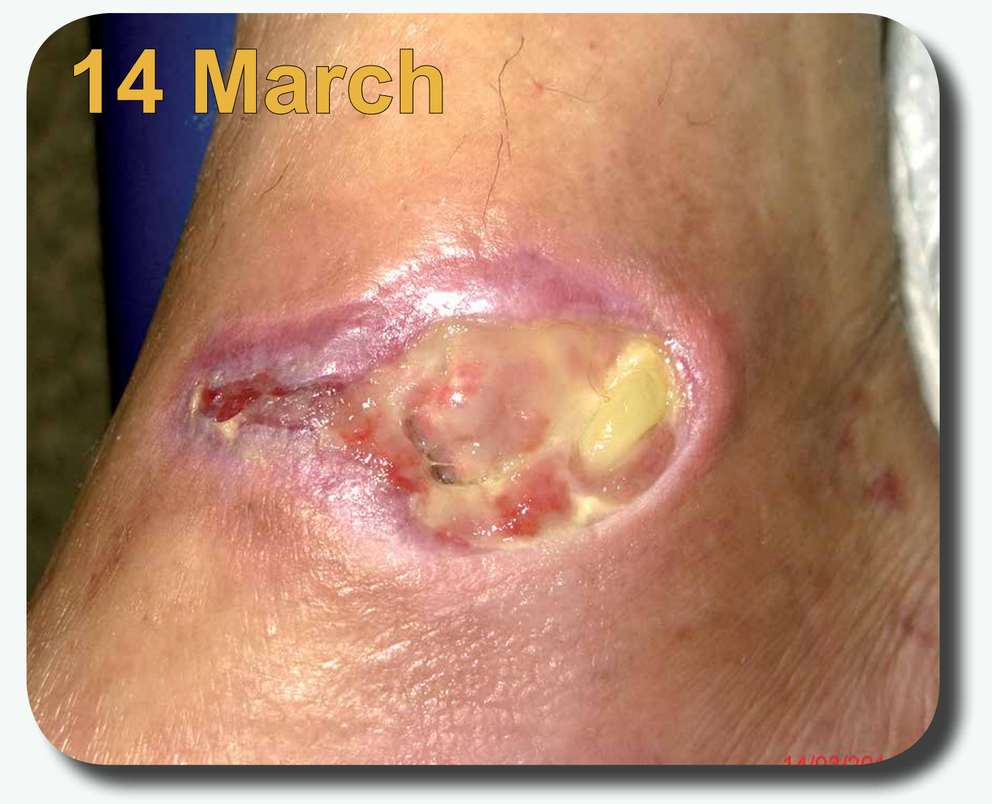
14 March

The surfactant reduces surface tension and glycerol helps maintain the moisture level of the wound; together promoting and enhancing a continuous cleansing and debriding of the wound in a painless fashion.

In spite of his poor prognosis and reduced peripheral circulation this wound started to heal.

An 88 year old male with Chronic Obstructive Airway Disease and a huge sacral pressure ulcer caused by spending most of his days sitting upright (to prevent dyspnea). A wound culture showed that the wound was infected with pseudomonas. Silver PMDs moistened with 3 ml normal saline solution were used to hydrate the necrotic tissue, and reduced the infection. No oral antibiotics were prescribed. Pain score 9 at initial assessment.

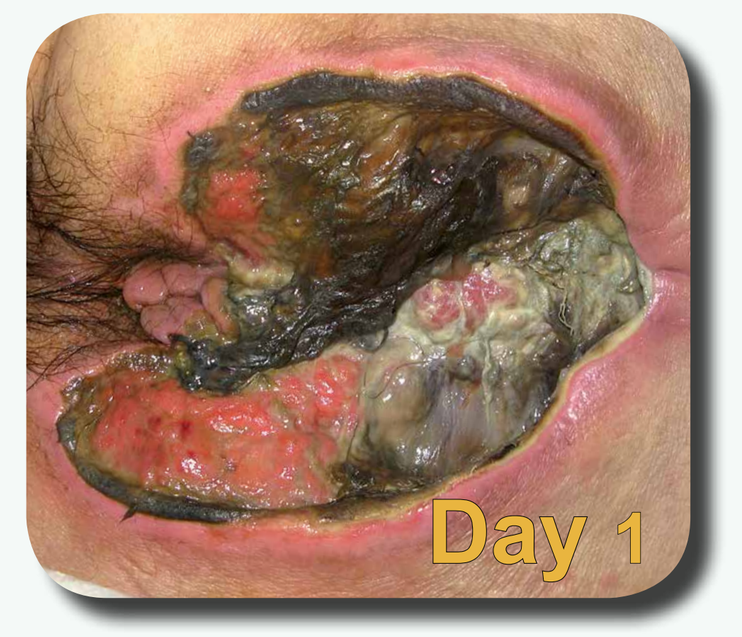
Day 1

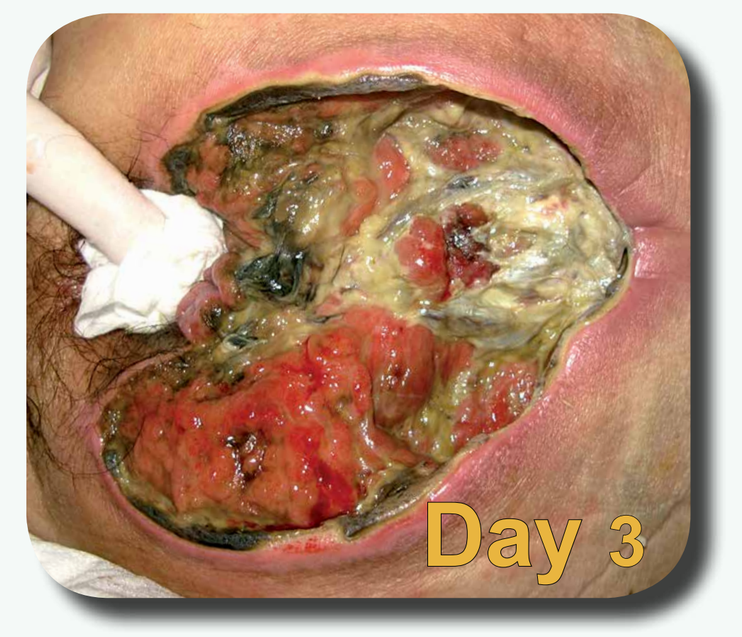
Day 3

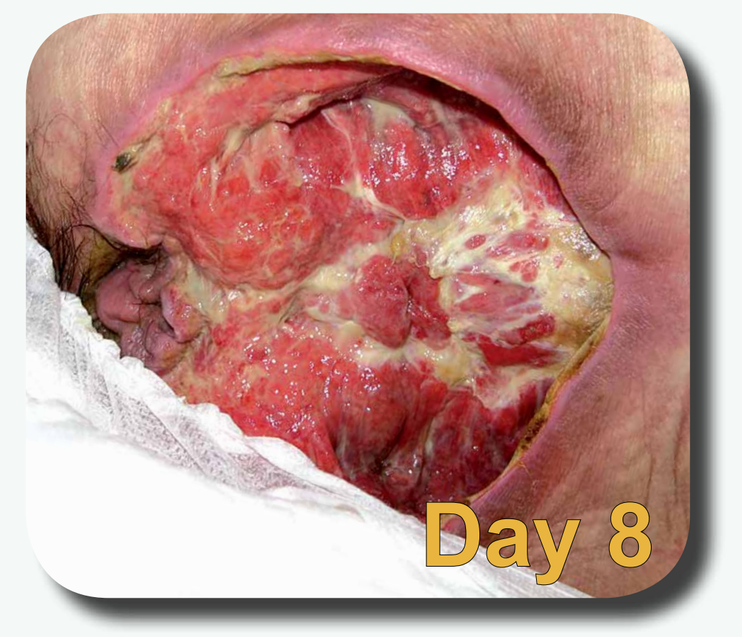
Day 8

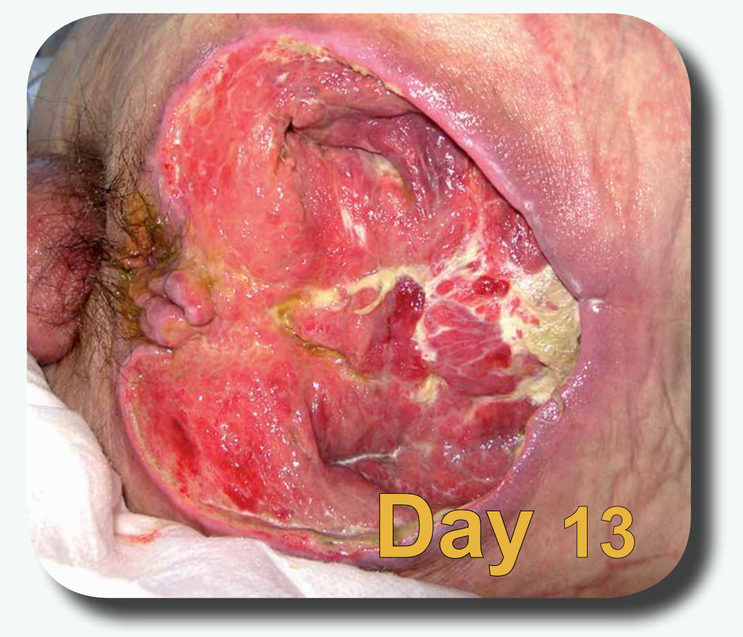
Day 13

The surfactant (wound cleanser) and glycerol incorporated in the dressing continually help cleanse and debride the wound. The hygroscopic action of glycerin cleans the wound by removing discharge with its osmotic action; reducing edema by inhibiting the lymphatic extension of infection. Pain level dropped to a 4 after two days.

Constant reinforced autolytic debridement of the wound leaves the wound bed, soft, clean, moisturized and smooth, with the surfactant reducing the bonding forces between the wound and necrotic tissue.

Pain score dropped to a level of 2 after a week's use of PMDs.

A 96 year old woman with Alzheimer's disease with an extensive pressure ulcer. Initial partial surgical debridement was done in the hospital prior to admittance to our center. Due to extensive bleeding the surgical debridement had to be stopped. She is on antibiotics, Levofloxacin and Amoxicillin.

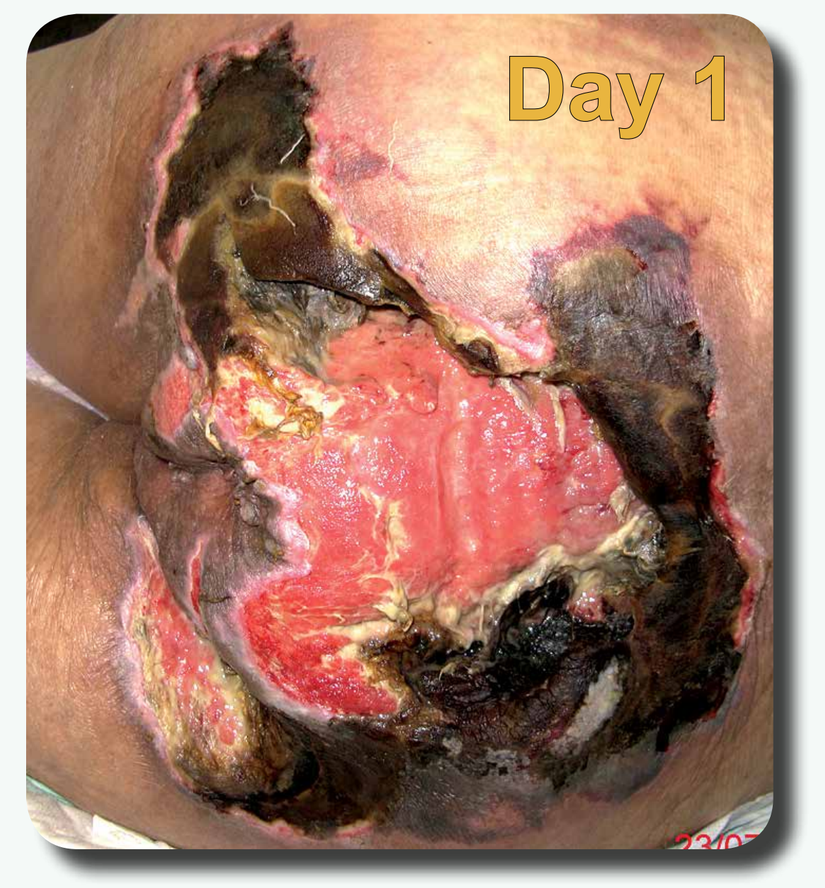
Day 1

PMD is used as a debriding agent. The wound is covered with a thick version of PMD. Over the necrotic tissue we moistened the PMD with a few drops of sterile water or saline in order to enhance the debridement process.

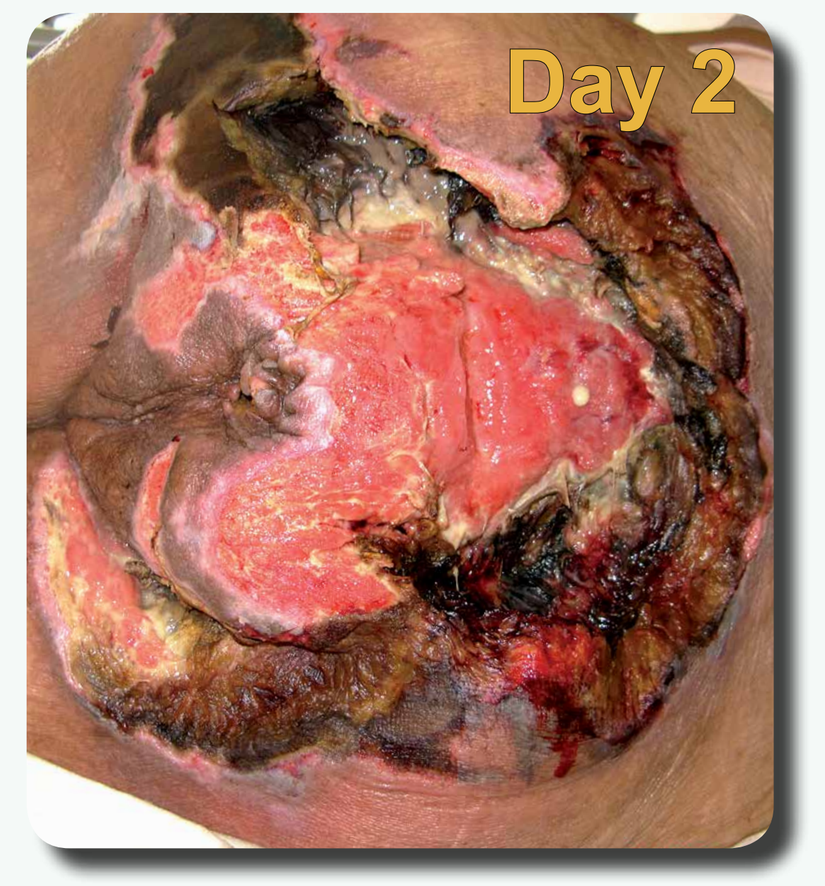
Day 2

During dressing change a PMD is gently rubbed over the necrotic tissue in a circular motion to enhance the autolytic debridement with a gentle mechanical debridement.

Pressure relieving mattress and repositioning every 2 hours.

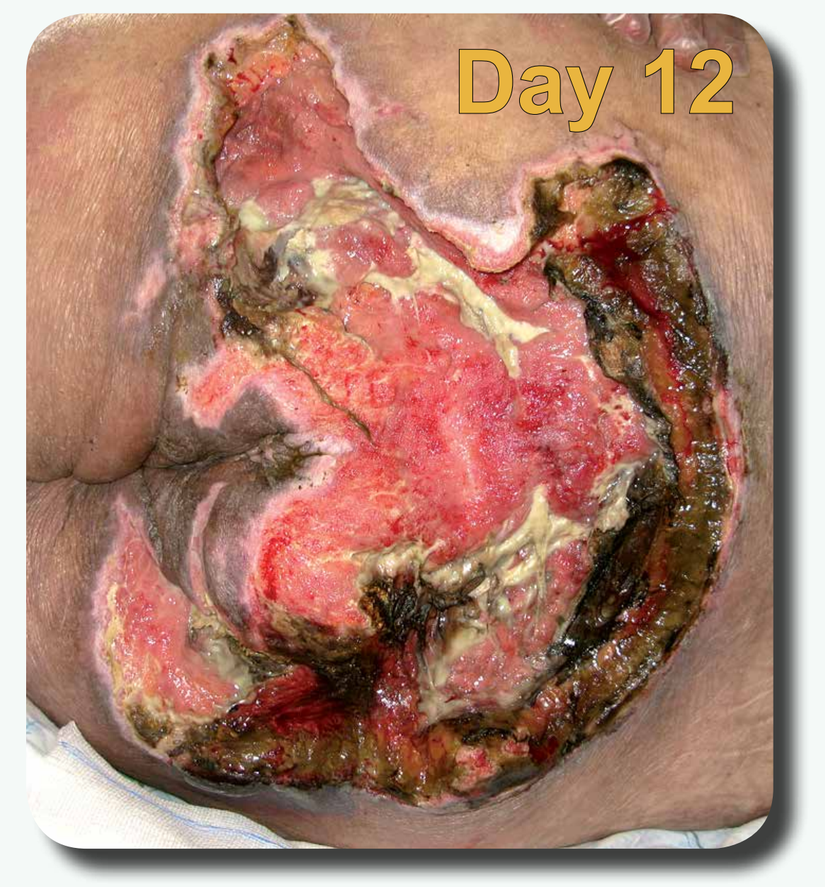
Day 12

Continuous autolytic debridement and healing is evident. The glycerin embedded in PMDs maintains an ideal moisture balance for wound healing; the dressing absorbs exudates and prevents maceration of the periwound area, reduces the interfacial tension between the viable and non-viable tissue, does not adhere to the wound bed, and protects it from further trauma.

A 92 year male, diabetic, with a pressure ulcer infected with MRSA. Previous treatment: i.v. antibiotics and hyaluronic acid cream have had no effect on this deteriorating wound.

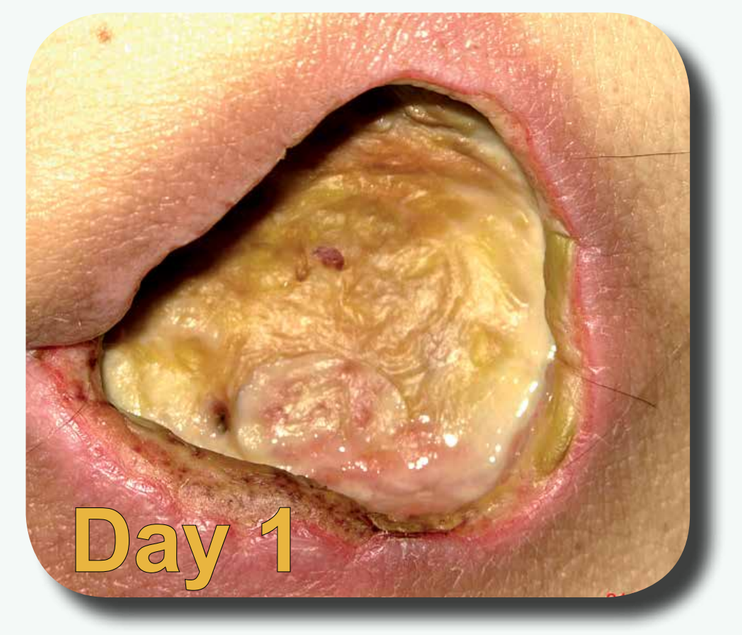
Day 1

Infected by MRSA after surgical debridement in the hospital. The patient's pain score was 8 out of 10. The dry sloughy wound was covered by a Silver PMD moistened with 3 ml saline.

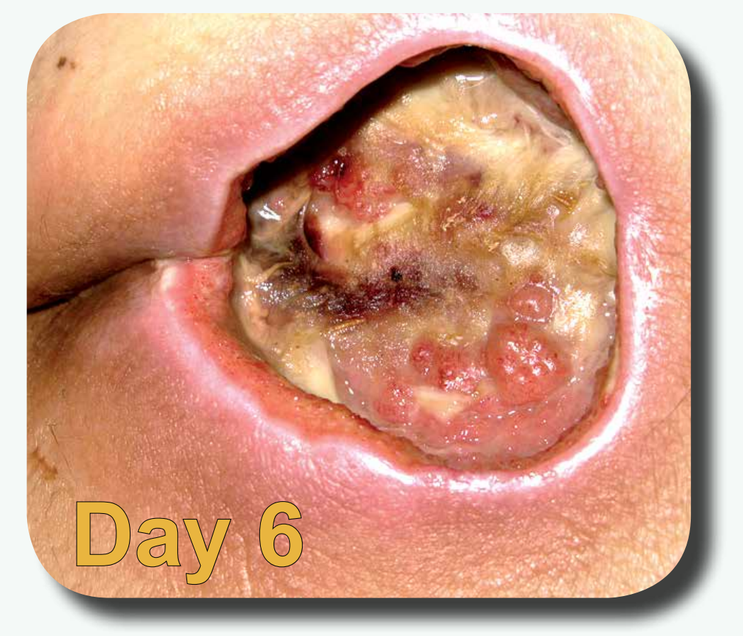
Day 6

Evidence of autolytic debridement. Pain score reduced to 3. The patients' albumin level was very low and we believe that the availability of glycerin within the PMDs allows it to act as a nutrient and energy substrate, creating a more optimal healing environment that is otherwise almost impossible to achieve in Diabetic patients.

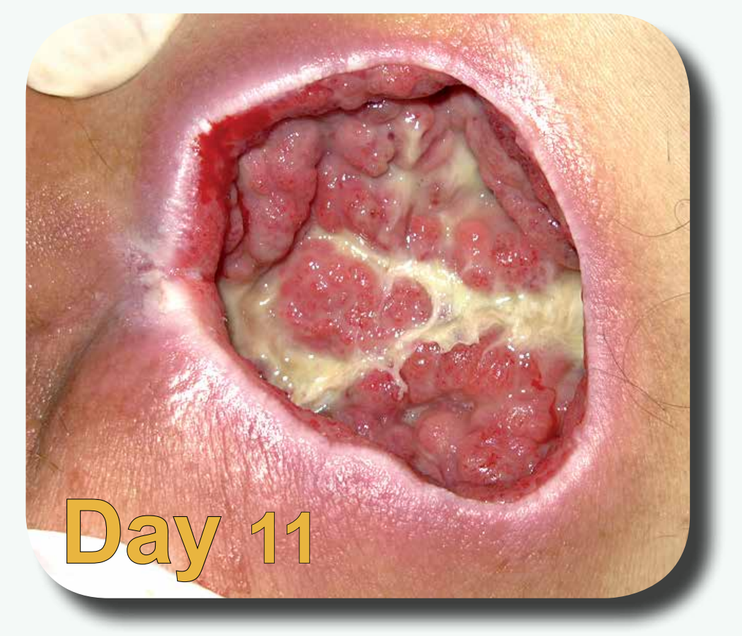
Day 11

The patient reports no pain at all. The wound is healing nicely and the exudate level is increasing. We no longer need to pre-moisten the dressing prior to application.

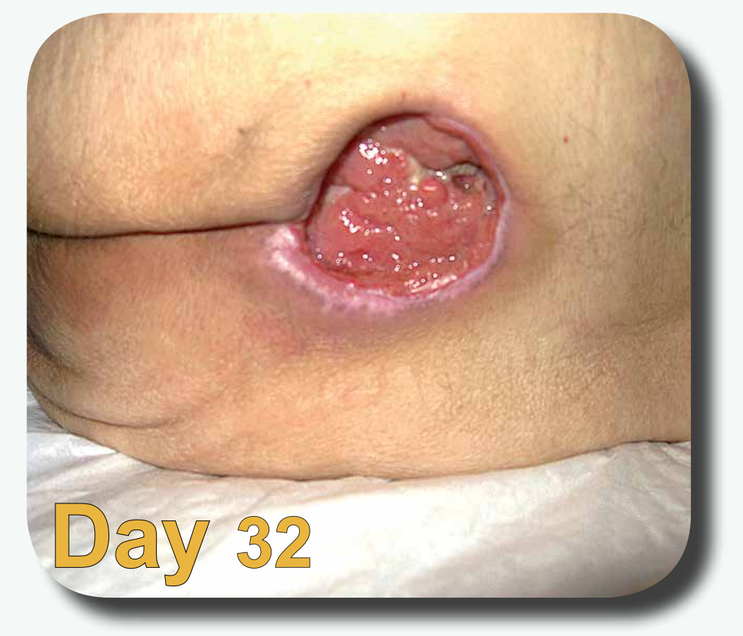
Day 32

Continuous healing and removal of the yellow slough. Cultures were taken and the wound was not infected with MRSA anymore so silver dressings no longer needed. We changed to the regular pink version of the PMD.

References

1. Beitz AJ, Newman A, Kahn AR, Ruggles T, Eikmeier L. A polymeric membrane dressing with antinociceptive properties: analysis with a rodent model of stab wound secondary hyperalgesia. The Journal of Pain. Feb 2004;5(1):38-47
2. Waller JM, Maibach HI (2006), Age and Skin structure and function, a quantitative approach (ii): protein, glycisoaminoglycans, water and lipid content and structure. Skin Research Technology, 12(3) pp 145-154
3. Rodeheaver GT, Smith SL, Thacker JG, Edgerton MT, Edlich RF. Mechanical cleansing of contaminated wounds with a surfactant. Am J Surg 1975; 129(3):241-5

*PolyMem® Wound Dressings with and without Silver. Manufactured by Ferris Mfg. Corp. 5133 Northeast Parkway, Fort Worth, TX 76106, USA . This case series was unsponsored.